Glembatumumab Vedotin, an Anti-gpNMB Antibody-Drug Conjugate (ADC), in Combination with Varlilumab in Patients with Advanced Melanoma

Abstract # P260

Omid Hamid[1], Anna C. Pavlick[2], C. Lance Cowey[3], Lowell Hart[4], Douglas B. Johnson[5], Jose Lutzky[6], Aaron Alizadeh[7], David Spigel[8], Neal Rothschild[9], April Salama[10], Robert Weber[11], Jason L. Luke[12], Ying Wang[13], Michael Yellin[13], Yi He[13], Abdel Halim[13], Thomas Hawthorne[13], Biwei Zhao[13], Rebecca G. Bagley[13], and Patrick A. Ott[14]

[1]The Angeles Clinic and Research Institute, Los Angeles, CA; [2]New York University School of Medicine, New York, NY; [3]Baylor University Medical Center, Dallas, TX; [4]Florida Cancer Specialists, Fort Myers, FL; [5]Vanderbilt University Medical Center and Vanderbilt Ingram Cancer Center, Nashville, TN; [6]Mount Sinai Comprehensive Cancer Center, Miami Beach, FL; [7]Northside Hospital, Atlanta, GA; [8]Tennessee Oncology, PLLC, Nashville, TN; [9]Florida Cancer Specialists, West Palm Beach, FL; [10]Duke University, Durham, NC; [11]St. Mary's Medical Center, San Francisco, CA; [12]University of Chicago, Chicago, IL; [13]Celldex Therapeutics, Inc., Hampton, NJ; [14]Dana-Farber Cancer Institute, Boston, MA

BACKGROUND

Glembatumumab vedotin (GV)

- Glycoprotein NMB (gpNMB):
 - Internalizable transmembrane glycoprotein overexpressed in multiple tumor types including ~80% of melanomas[1]
 - High tumor gpNMB expression associated with shorter metastasis-free and overall survival[2-4]
 - Upregulated in $BRAF^{V600}$ mutant melanoma following BRAF/MEK inhibition[5]
- GV is a gpNMB-targeting antibody, CR011, linked to the potent cellular toxin monomethylauristatin E (MMAE) using Seattle Genetics' proprietary technology
 - Delivers MMAE to gpNMB-expressing tumor cells

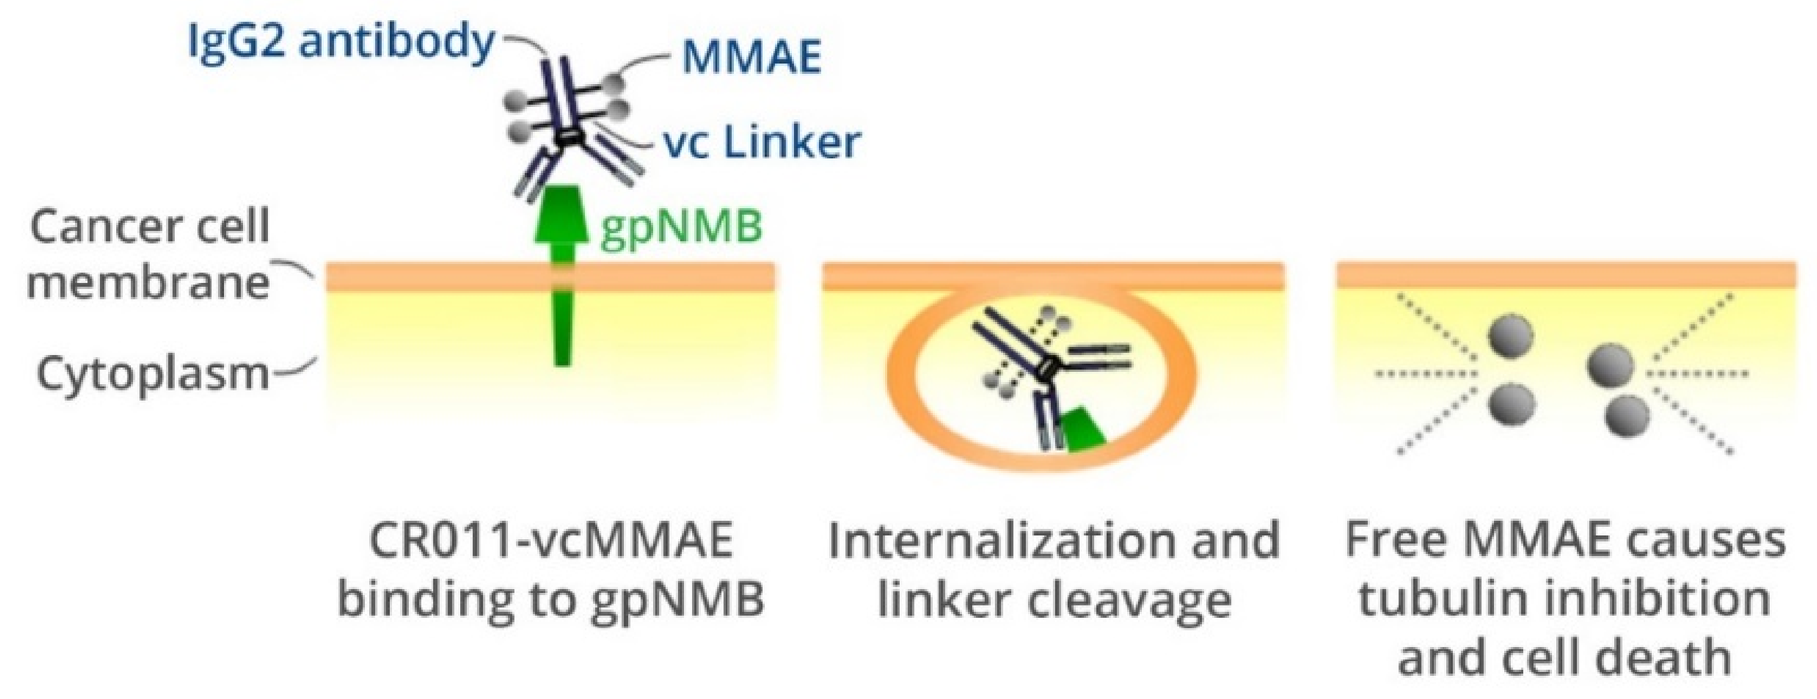
IgG2 antibody
MMAE
vc Linker
Cancer cell membrane
gpNMB
Cytoplasm
CR011-vcMMAE binding to gpNMB
Internalization and linker cleavage
Free MMAE causes tubulin inhibition and cell death

- Preclinical data suggest synergistic anti-tumor activity when ADC-MMAE is combined with checkpoint inhibitors (CPI)
- Microtubule-depolymerizing cytotoxic agents (e.g., MMAE) have been shown to convert tumor-resident tolerogenic dendritic cells into active antigen-presenting cells[6, 7]

Varlilumab

- CD27
 - Member of the TNF-receptor superfamily
 - Constitutively expressed on most T cells and a subset of B and NK cells
 - CD27 signaling:
 - Activation of the NF-κB pathway
 - Cell survival, activation, proliferation
 - Role in generation and long-term maintenance of T cell immunity
 - Role in NK cell differentiation and activation

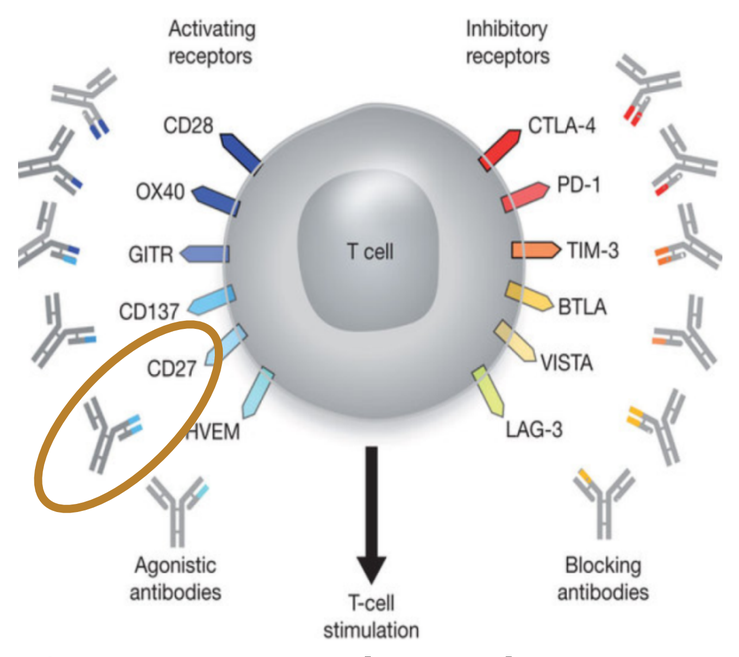
Activating receptors
Inhibitory receptors
CD28
OX40
GITR
CD137
CD27
HVEM
T cell
CTLA-4
PD-1
TIM-3
BTLA
VISTA
LAG-3
Agonistic antibodies
T-cell stimulation
Blocking antibodies

- Varlilumab is a fully human IgG_1 CD27 agonist monoclonal antibody
 - Induces activation and proliferation of human T cells when combined with T cell receptor stimulation
 - Strong preclinical data demonstrating single agent and combination activity in tumor models
 - Safety, biological and clinical activity demonstrated in Ph 1 study[8]
 - Safety and tumor infiltrating lymphocytes increase demonstrated in combination with nivolumab in Ph 1 study[9]

CDX011-05 STUDY DESIGN

Study Design

- Single arm, open-label, Ph 2 trial
- Sequential cohorts evaluating GV (1.9 mg/kg IV q3w) as monotherapy or in combination with immunotherapy
- Tumor assessments every 6 weeks for 6 months, then every 9 weeks

Patient (Pt) Population

- Unresectable Stage III or IV melanoma
- Refractory to checkpoint inhibition (anti-CTLA-4, -PD-1 or -PD-L1)
- Refractory to BRAF/MEK inhibition (if $BRAF^{V600}$ mutant)
- ≤1 prior cytotoxic regimen

Endpoints / Statistical Design

Primary

- Objective response rate (RECIST 1.1)

Secondary

- Progression free survival, duration of response, overall survival, safety, and tumor gpNMB expression vs. outcome

Exploratory

- Potential biomarkers and pharmacokinetics

Cohort 1: GV Monotherapy

- Encouraging activity in pts with advanced melanoma refractory to CPI[10]
- Primary study endpoint of ORR was met:
 - ORR = 11% (95% CI: 4.7, 21.9)
 - Duration of response = 6.0 months (95% CI: 4.1, NR)
 - 52% pts experienced disease control (SD or better ≥3 months)

Cohort 2: GV with Varlilumab

- Varlilumab: 3.0 mg/kg IV on Day 1 of Weeks 1, 3, 9, 15, 21 and 27
- 34 pts treated
- 2 pts continue on treatment; 14 pts remain in survival follow-up
- Designed to estimate overall response rate
 - Not powered for statistical comparisons between cohorts

PATIENT CHARACTERISTICS

Demographics and Disease Characteristics (n=34)	
Male *(n [%])*	20 (59)
Age, years *(median [min, max])*	61 (30, 86)
ECOG PS 0-1 *(n [%])*	34 (100)
Stage *(n [%])*	
III	1 (3)
IV	33 (97)
M1a	2 (6)
M1b	4 (12)
M1c	27 (79)
Duration of Advanced Disease, months *(median [min, max])*	22.3 (2.3, 199.6)
BRAF Mutation *(n [%])*	9 (27)
≥ 4 Disease Sites *(n [%])*	11 (32)
Prior Anti-Cancer Regimens *(median [min, max])*	3 (1, 8)
Prior Therapies *(n [%])*	
Checkpoint Inhibitor	34 (100)
# Prior CPI *(median [min, max])*	2 (1, 4)
Anti-CTLA-4	26 (76)
PD-1/PD-L1 Inhibitor	34 (100)
BRAF or BRAF/MEK Inhibitor	11 (32)
Chemotherapy	10 (29)
Cytokines	14 (41)

EXPOSURE

Exposure (n=34)	
Doses of GV *(median [min, max])*	3 (1, 12)
Doses of Varlilumab *(median [min, max])*	2 (1, 10)
Duration of GV Exposure, days *(median [min, max])*	46.5 (1.0, 253.0)
Duration of Varlilumab Exposure, days *(median [min, max])*	26.0 (1.0, 212.0)

GV Serum Concentrations (n=19)

Methods
- Serum: baseline, Cycles 1-2, end of treatment
- Total antibody (TA): antigen capture/IgG detection (ELISA)
- ADC: anti-MMAE capture/antigen detection (ELISA)
- Free MMAE: LC/MS-MS

Results
- No indication of analyte accumulation over this schedule
- The addition of varlilumab did not appear to influence the exposure of the GV conjugate, TA, or MMAE as compared to GV monotherapy

TOLERABILITY

Adverse Events Considered Related to GV and/or Varlilumab (n=34)

	Overall	Grade ≥3 Severity (CTCAE)
Fatigue	56%	9%
Rash	53%	21%
Pruritus	50%	3%
Alopecia	35%	NA
Neuropathy	26%	3%
Diarrhea	26%	0
Nausea	24%	0
Decreased Appetite	24%	0
Vomiting	21%	0
Neutropenia	18%	12%
Anemia	15%	9%
Leukopenia	15%	9%
AST Increased	12%	0

Table includes toxicity assessed as related to GV and/or varlilumab occurring at any severity in ≥10% of pts

- 1 pt discontinued: Grade 4 cellulitis at skin biopsy site
- No treatment-related death

References

1. Tse et al. CCR 2006
2. Rose et al. CCR 2010
3. Li et al. APMIS 2013
4. Kuan et al. CCR 2006
5. Rose et al. CCR 2016
6. Muller et al. CIR 2014
7. Muller et al. STM 2015
8. Burris et al. JCO 2017
9. Sanborn et al. JCO 2017
10. Ott et al. JCO 2017

ACTIVITY

Clinical Efficacy

Primary Endpoint: Confirmed Responses (ORR)	1/31# (3%)
Any Response Including Those Not Confirmed at Subsequent Assessment	3/31 (10%)
Stable Disease*	16/31 (52%)
Disease Control Rate (DCR)**	6/31 (19%)
Patients with Tumor Shrinkage	16/31 (52%)

3 pts excluded from response-evaluable population due to no post-baseline assessments (unrelated AE, consent withdrawals)
* Stable disease with minimum interval ≥6 weeks from baseline
** Includes pts without progression for >3 months

Progression Free Survival (PFS)

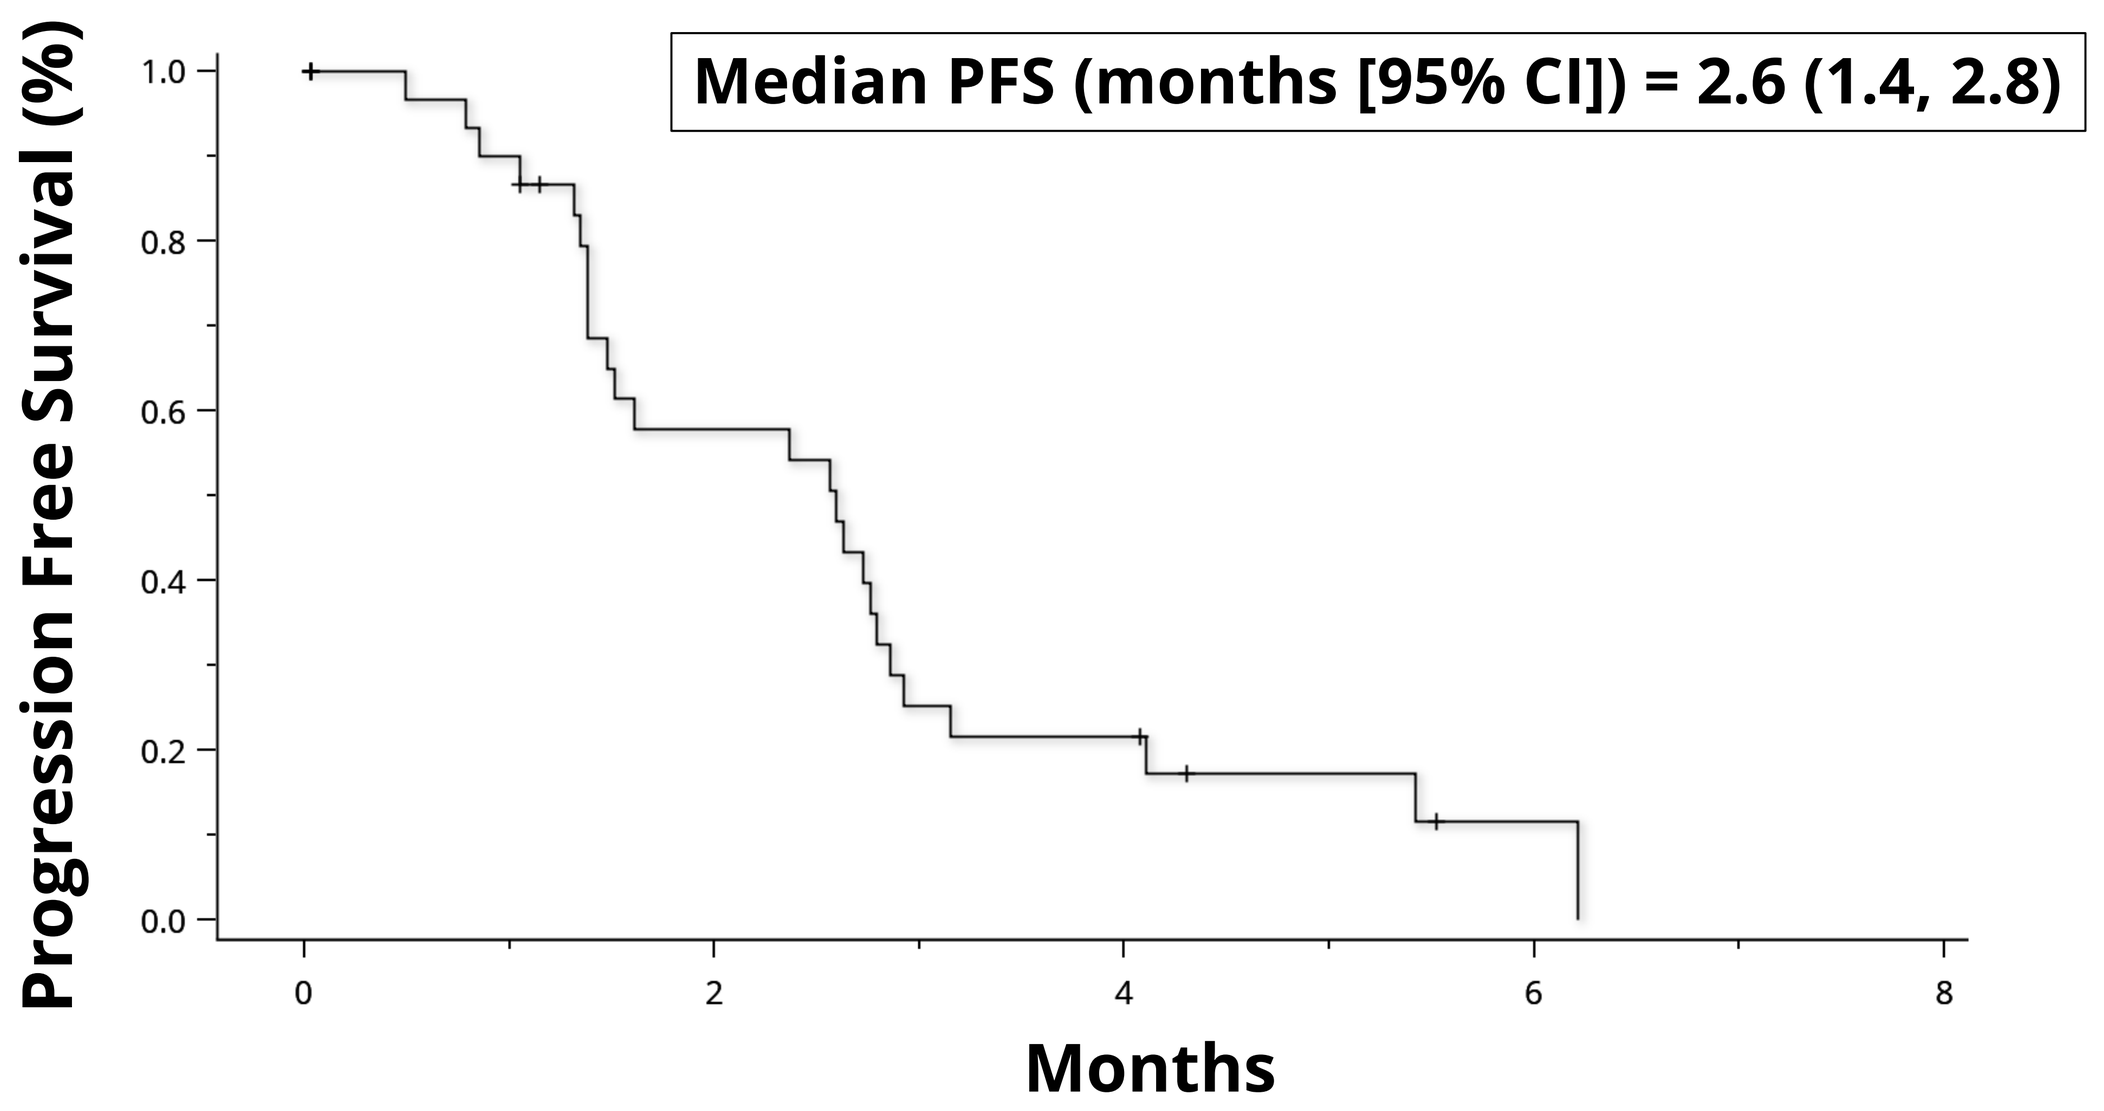
Median PFS (months [95% CI]) = 2.6 (1.4, 2.8)
Progression Free Survival (%)
1.0
0.8
0.6
0.4
0.2
0.0
0
2
4
6
8
Months

Maximum Tumor Shrinkage

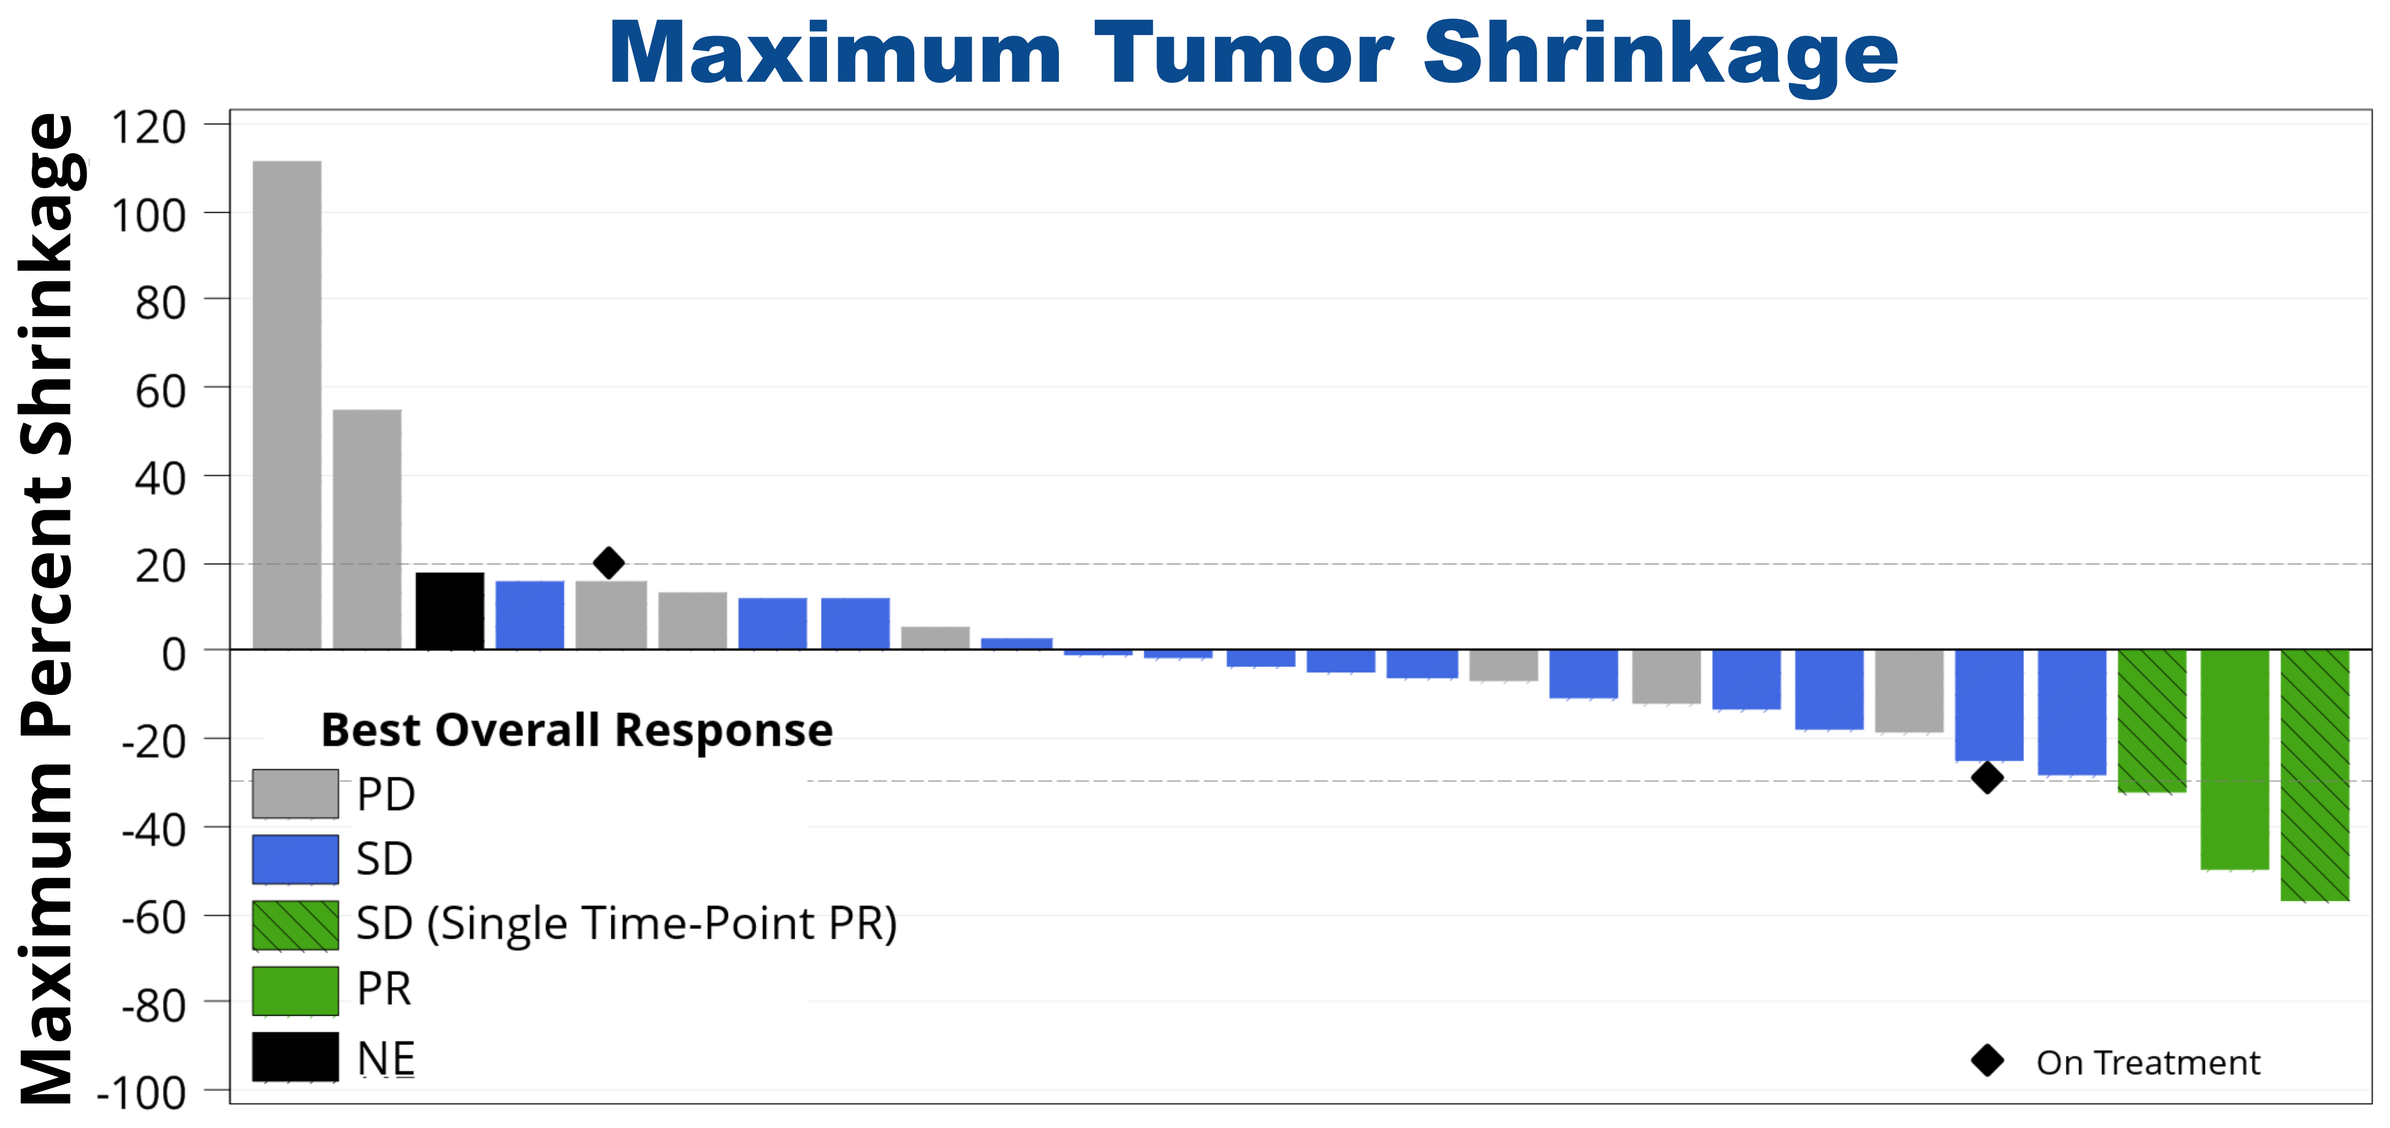
Maximum Percent Shrinkage
120
100
80
60
40
20
0
-20
-40
-60
-80
-100
Best Overall Response
PD
SD
SD (Single Time-Point PR)
PR
NE
On Treatment

Overall Survival (OS)

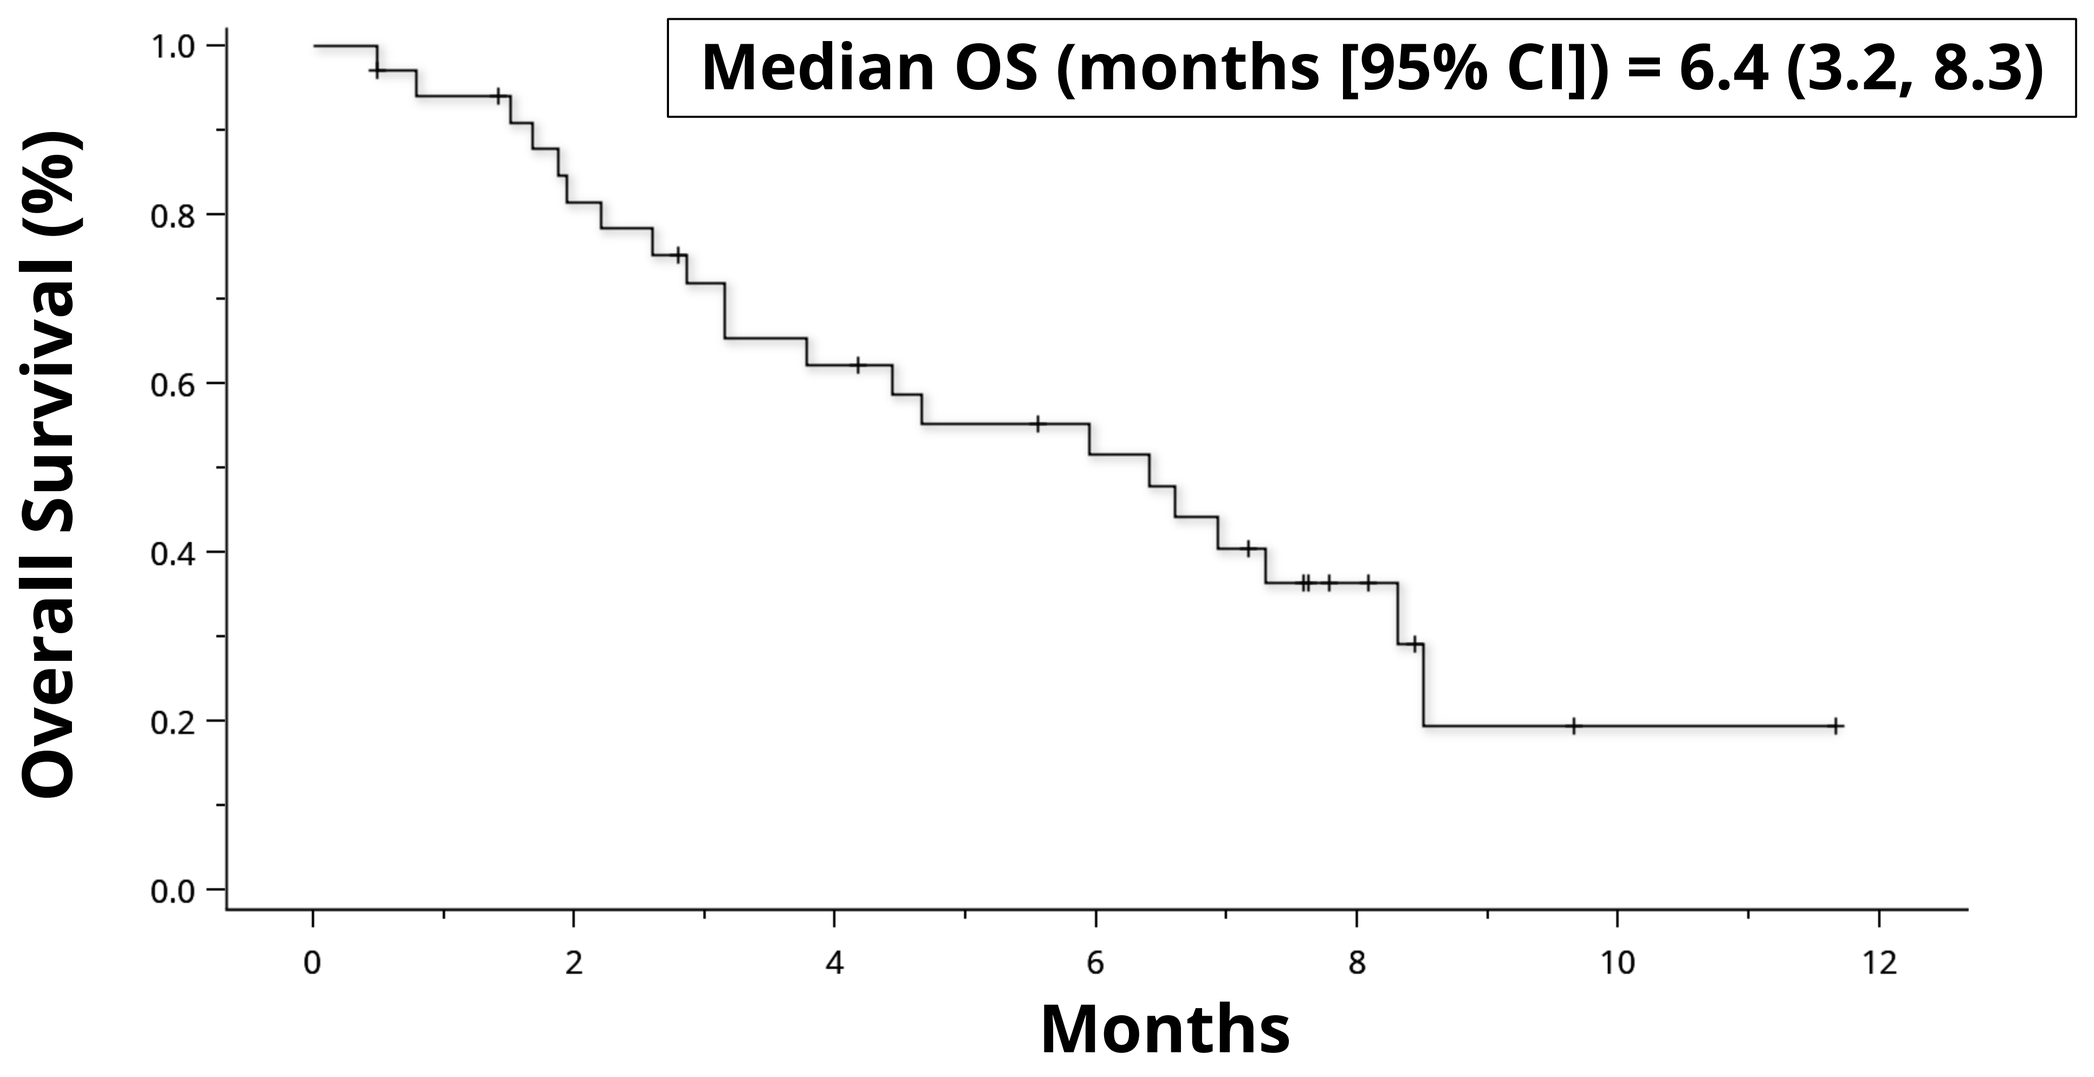
Median OS (months [95% CI]) = 6.4 (3.2, 8.3)
Overall Survival (%)
1.0
0.8
0.6
0.4
0.2
0.0
0
2
4
6
8
10
12
Months

TRANSLATIONAL RESEARCH

Flow Cytometry Analysis of Immune Cell Subsets

Effects of varlilumab on peripheral lymphocytes are consistent with prior studies of varlilumab

- Activation of T cells: evidenced by up-regulation of HLA-DR (P=0.08)
- Decrease in circulating regulatory T cells (Treg) (P=0.003)

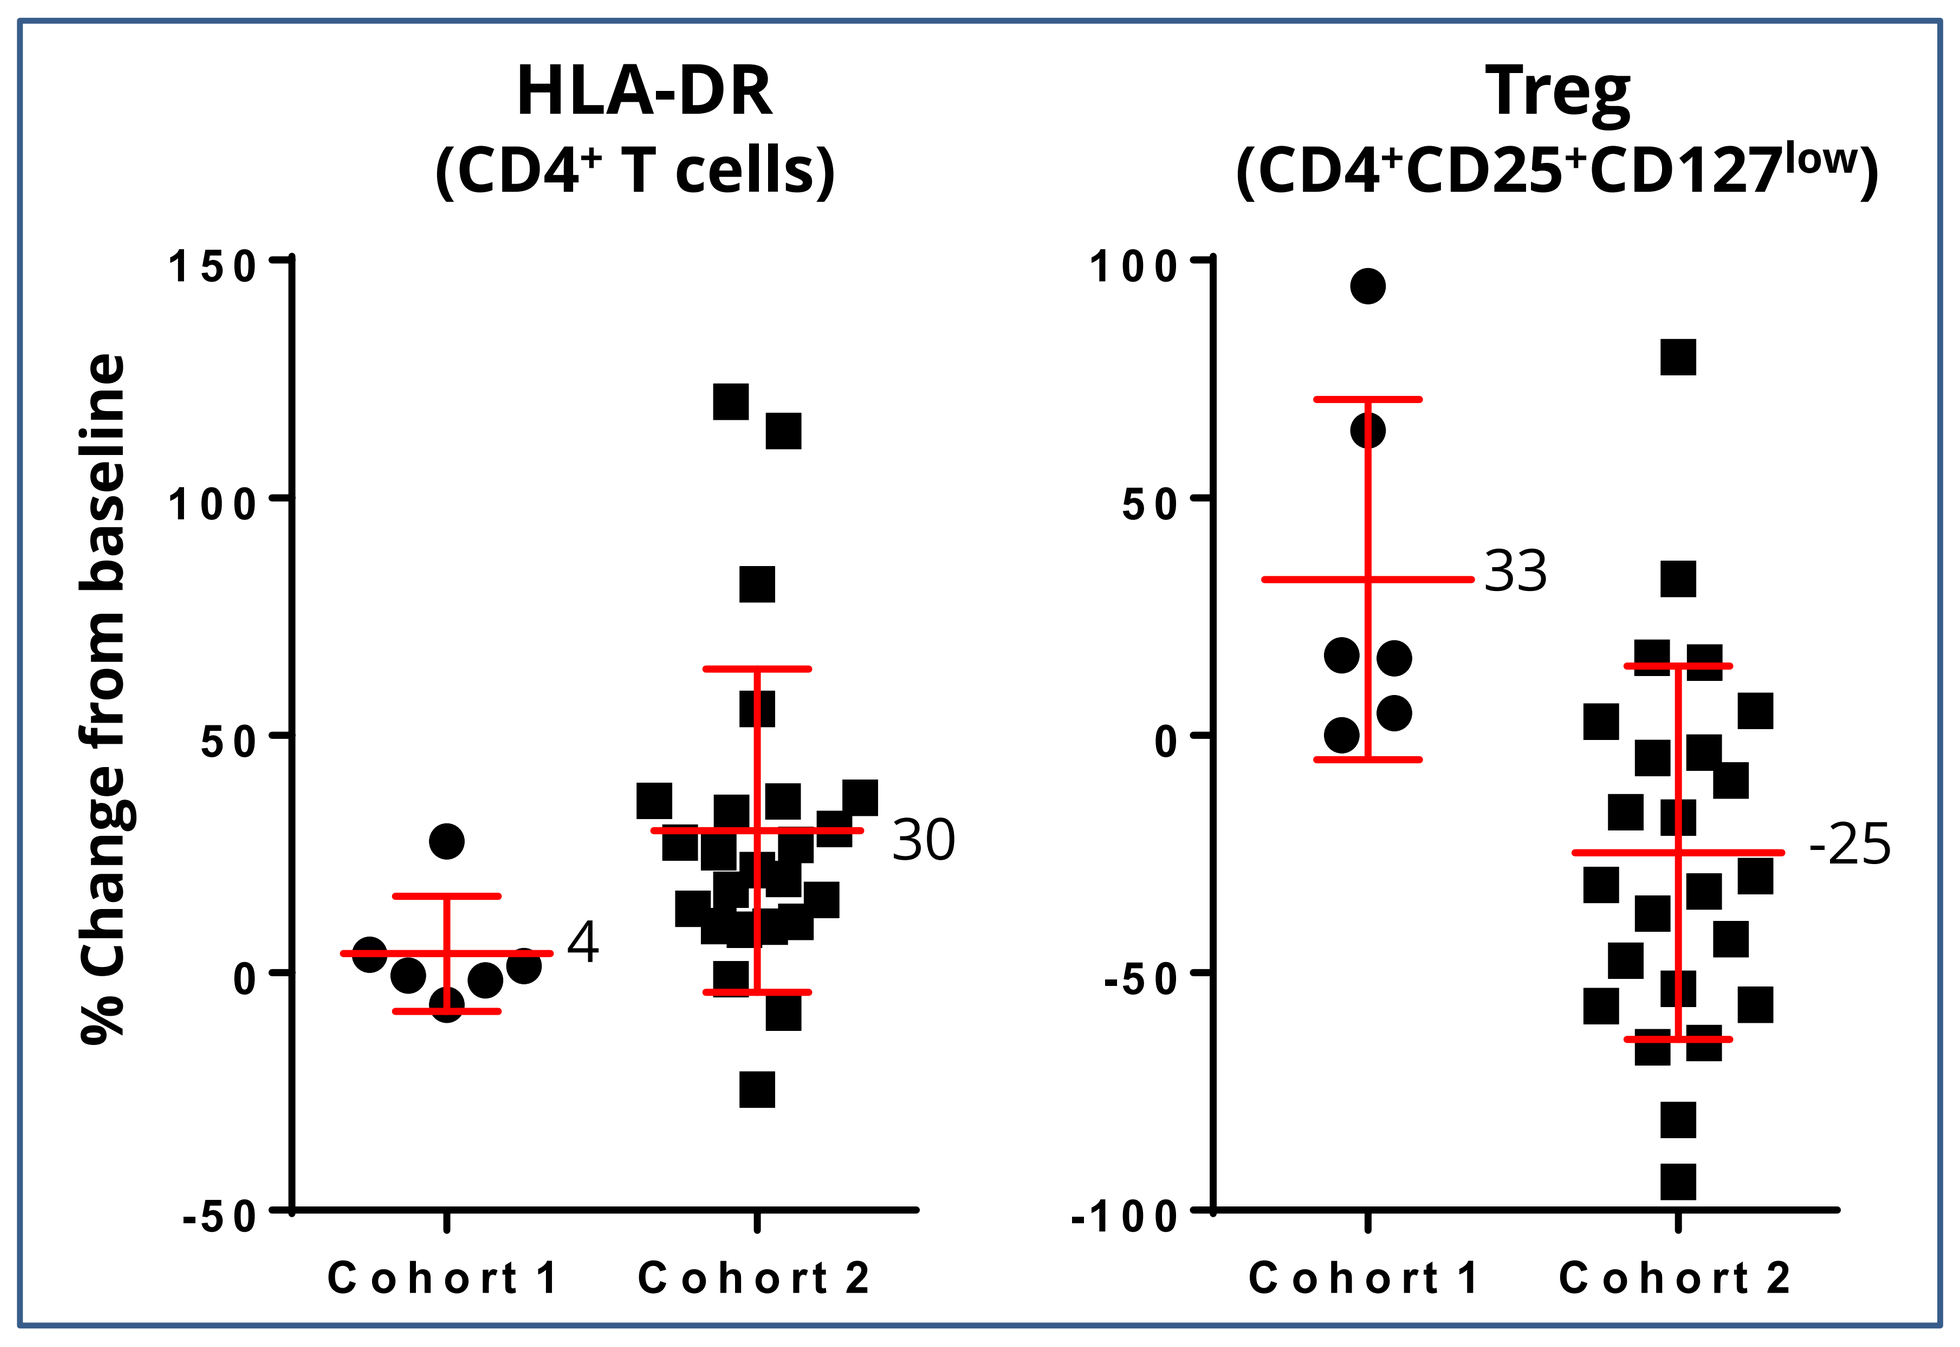
HLA-DR
($CD4^+$ T cells)
Treg
($CD4^+CD25^+CD127^{low}$)
% Change from baseline
150
100
50
0
-50
-100
4
30
33
-25
Cohort 1
Cohort 2

Flow cytometry analysis performed on purified PBMC (stored frozen until analysis). Baseline and end of cycle 1 samples were evaluated for % positive cells and plotted as % change from baseline. Six randomly selected pts from Cohort 1 were selected for comparison.

Correlative Analyses

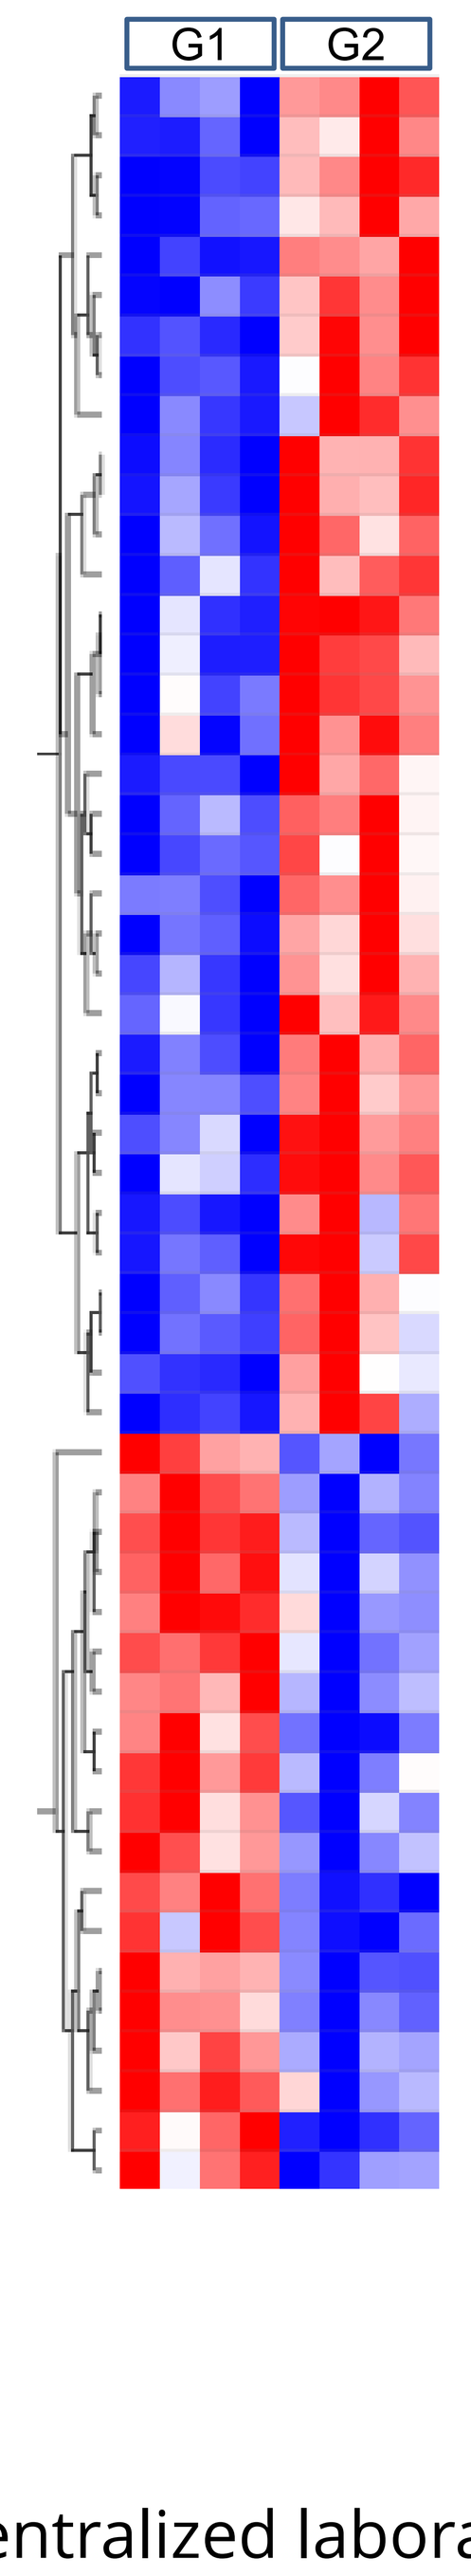
G1
G2

- Prior GV studies, and Cohort 1, suggested a correlation between skin rash and clinical outcome.
- Pre-treatment fresh skin biopsies were collected to identify a gene signature that may predict rash and thus, outcome.
 - Skin biopsies: stored in RNALater, -70ºC; RNA extraction; QC; and analyzed on Agilent microarray: SurePrint G3 Human Gene Expression v3 8x60K Microarray Kit.
- Although no significant correlation between gene signature and rash, or rash and outcome was observed in Cohort 2, a secondary analysis was performed to investigate gene expression and outcome.
- To elucidate any possible differentially expressed genes (DEGs), data from the 4 best responders (tumor shrinkage > 20% and PFS > 3 months; G1) was compared with the 4 worst responders (tumor growth >10% and PFS <2 months; G2).
 - Fold change >2 & p <0.05: 198 DEGs
 - Fold change >2 & p <0.01: 76 DEGs
 - Fold change >2 & p < 0.005: 53 DEGs
 - Heatmap (left): 53 DEGs

These up- and down-regulated genes and relevant pathways are being evaluated for their potential role with regard to response to the gpNMB-targeted ADC.

Tumor gpNMB Expression

- Pre-entry tumor tissues analyzed by immunohistochemistry at a centralized laboratory
- Tumors were gpNMB+ for all 31pts with available tissue, and 87% had tumors with 100% epithelial cells gpNMB+
- Stable expression (65-100% gpNMB+) over time in primary vs. metastatic tumors (14 pts)
- Consistently high gpNMB expression did not allow for evaluation of correlation with outcome

CONCLUSIONS

- Glembatumumab vedotin/varlilumab was well tolerated without evidence of additive toxicity
- Biological effects of varlilumab were consistent with prior observations and did not appear to be impacted by the addition of an ADC
- No apparent enhanced clinical benefit by the addition of varlilumab to glembatumumab vedotin; possibly due to:
 - Potential lack of sensitivity to immunotherapy in patients with checkpoint refractory disease
 - Rapidly progressive, treatment-refractory pt population resulting in short duration of treatment (median 2 doses of varlilumab)
 - Possible dearth of antigen presenting cells in tumors
 - Immune checkpoint molecules remained unblocked
- High gpNMB expression in cohort 2 was similar to that observed in cohort 1
- Distinct molecular pattern in normal skin associated with improved outcome; further evaluation will explore the potential for patient selection in subsequent studies

FUTURE DIRECTIONS

- Building upon the positive monotherapy results, an additional cohort evaluating GV in combination with CPI is enrolling (NCT#02302339)
- Additionally, Flt3L is a growth factor for dendritic cells. CDX-301, a recombinant human Flt3L, has been tested in clinical trials in the oncology setting
- A subsequent cohort is planned to evaluate combination of GV with CDX-301 to assess the safety, tolerability, and biologic activity of the combination
- Future correlative analyses will include molecular profiling with NextGen sequencing on tumor tissues to investigate possible impact of genetic makeup on clinical outcome
- Following completion of this cohort and evaluation of available data, the protocol amendment also allows for the exploration of additional cohorts

Celldex therapeutics

View Poster